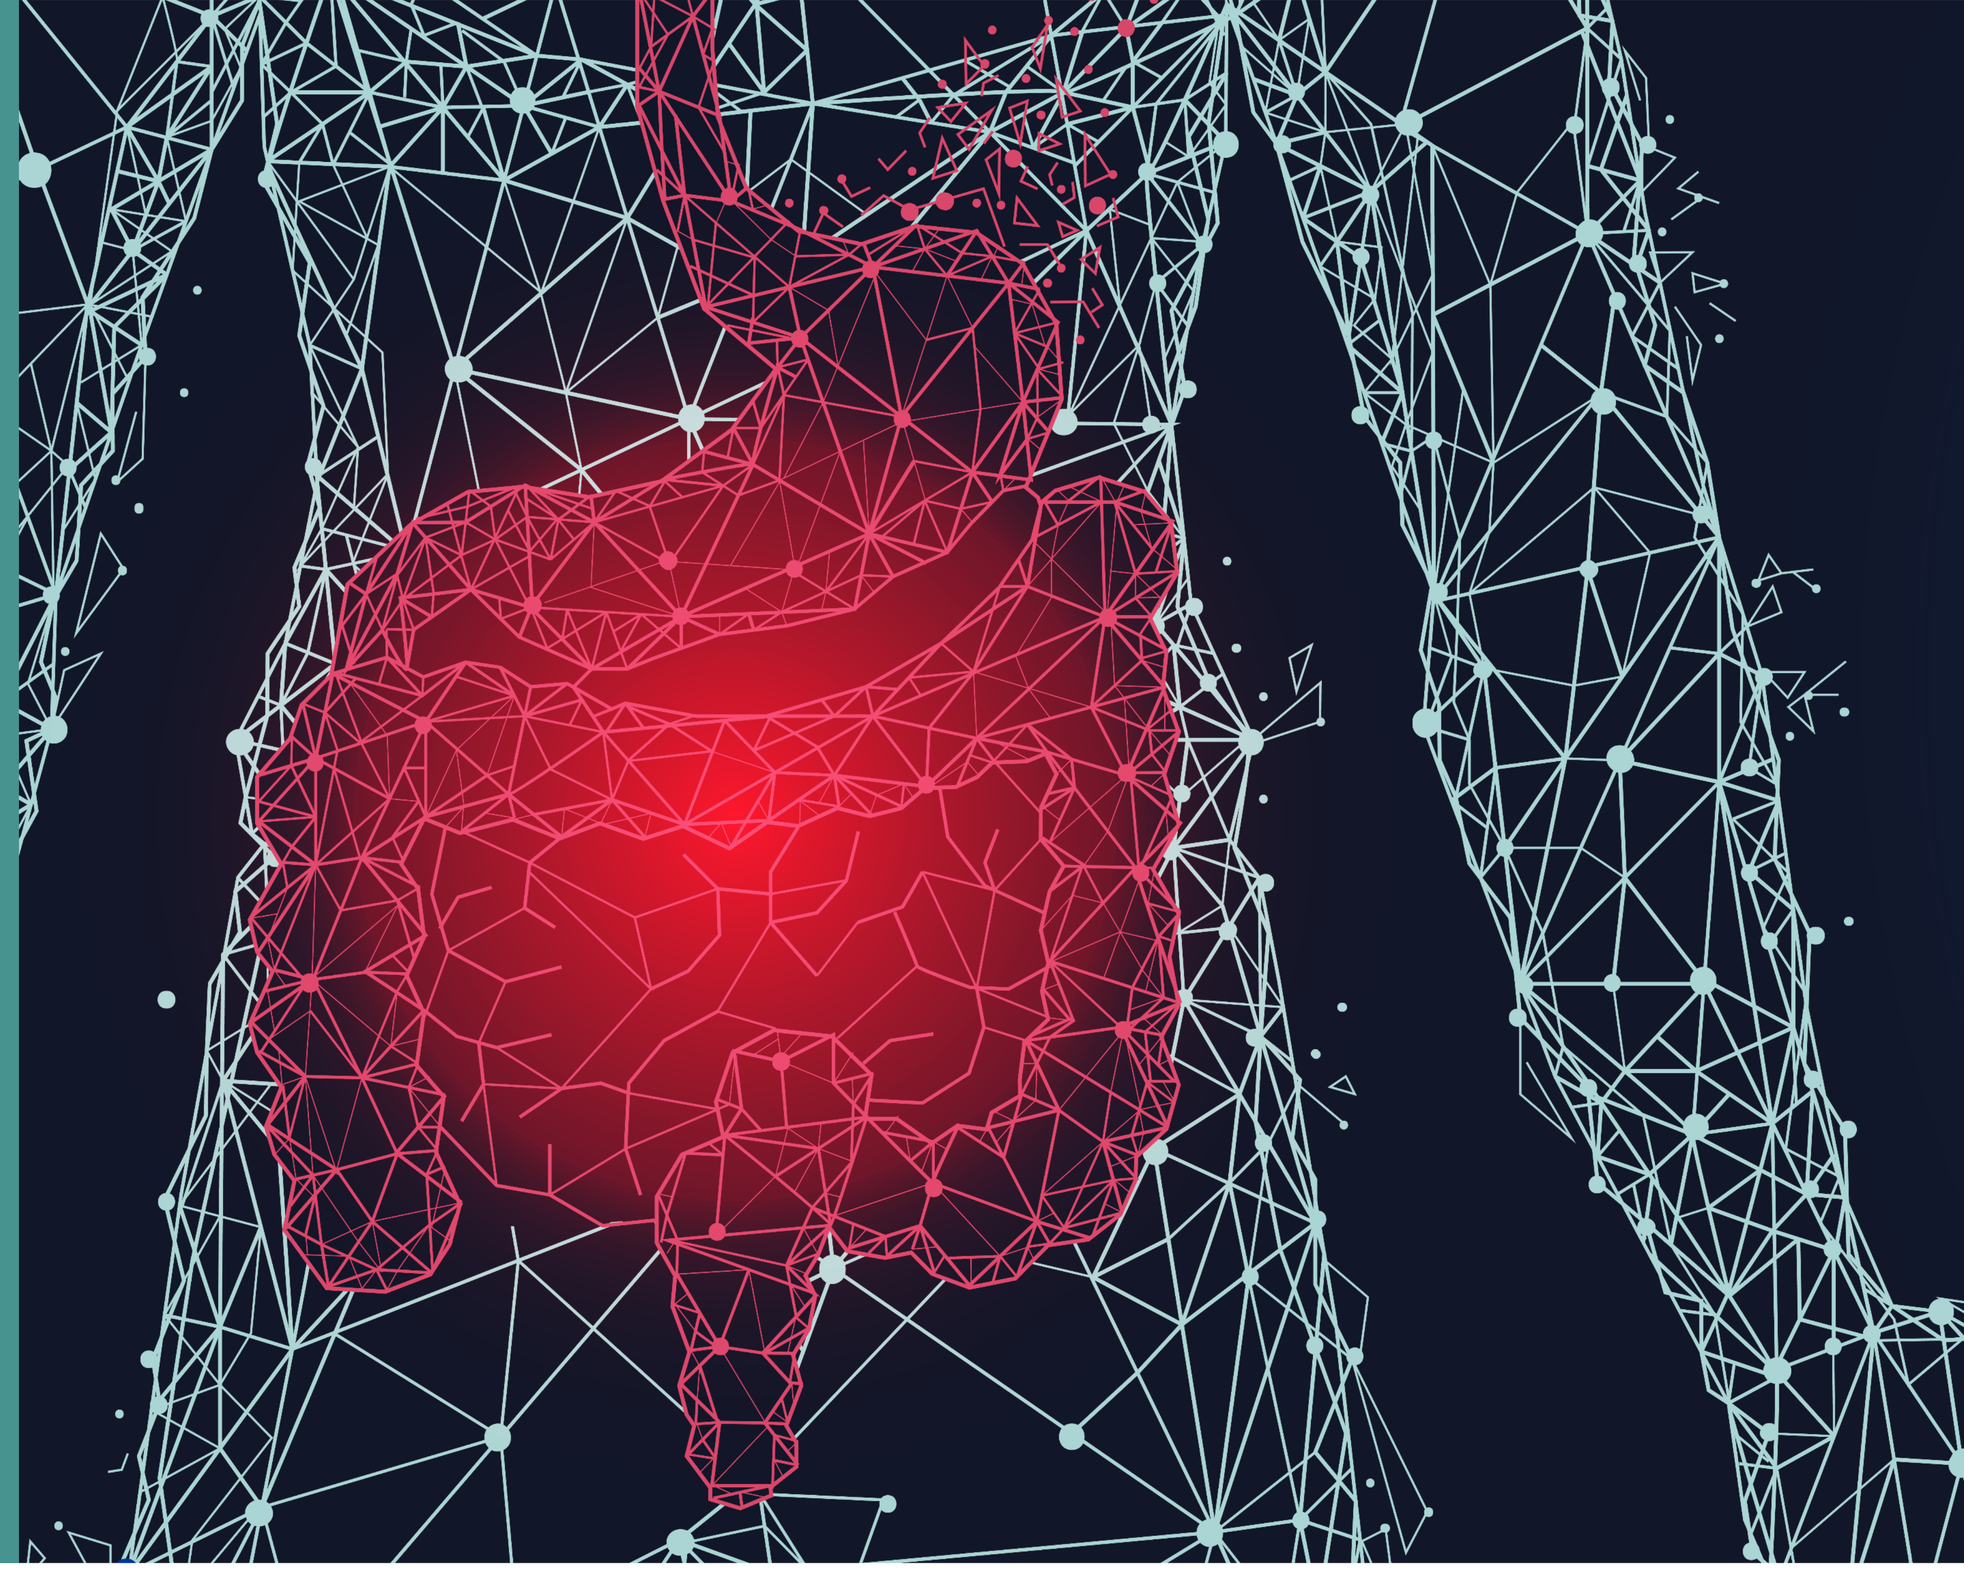

LOCAL BIOACTIVATION AND EFFICACY OF PALI-2108: A PROMISING PDE4 INHIBITOR PRODRUG FOR ULCERATIVE COLITIS TREATMENT

Joie Bernard[1] PhD, Christophe Mellon[2] PhD, Maxime Ranger[2], Bernard Cote[2] PhD, and Mitchell Jones[1] MD PhD

1. Palisade Bio, Carlsbad, CA. United States; 2. Giiant Pharma, Montreal, Quebec.

Introduction

Phosphodiesterase-4 (PDE4) is a key enzyme in cAMP hydrolysis and its inhibition elevates intracellular cAMP, downregulates inflammatory cytokines, and reduces the expression of cell adhesion molecules, thereby preventing local infiltration and activation of inflammatory cells. Approved PDE4 inhibitors include roflumilast for use in chronic obstructive pulmonary disease (COPD) and apremilast for psoriasis/psoriatic arthritis. Despite the development of subtype specific PDE4 inhibitors, oral administration and systemic distribution has resulted in central nervous system (CNS) toxicity, such as headaches, nausea or vomiting, leading to discontinuation of therapy and limiting potential efficacy. Targeted and better-tolerated oral PDE4 inhibitors hence remain an unmet need in IBD.

We investigated PALI-2108, an orally delivered, intestinally activated PDE4 inhibitor prodrug designed with a D-glucoronic sugar moiety. PALI-2108 minimizes systemic exposure until cleaved by the colonic bacterium enzyme β-glucuronidase, producing active drug PALI-0008, therefore reducing potential CNS toxicity. PALI-2108 proposed mechanism of action is outlined in figure 9.

Methods

Healthy mice were dosed with a single oral dose of PALI-2108 and sacrificed at multiple time points across 24 hours. Proximal colon, distal colon and plasma were collected and PALI-0008 measured (Figure 3).
Mice were treated once with 3% oxazolone i.r. to induce colitis, followed by treatment with PALI-2108 BID at 4.2 mg/kg for 3 days. PALI-2108,-0708, and -0008 were measured at 72 hours in plasma, duodenum, ileum and colon (Figure 4).
A classic cellular thermal shift assay (CETSA) was developed to assess on-target PDE4 binding within colon tissue homogenates dosed with apremilast, PALI-2108 or Vehicle and detecting changes in thermal stability (Figure 1 and 5).
A second acute colitis model of disease induction in mice was utilized with 2% DSS in drinking water administered from D0 to D7. Mice were treated on D1 through D6 with PALI-2108 BID, PALI-2108 1.5mg/kg, tacrolimus and apremilast were administered QD. Mice were sacrificed on day 7 (Figure 2). Disease activity index (DAI) score was assessed based on a scoring system including body weight loss, stool consistency and the presence of blood in feces (Figure 6 and 7).
PO administration of PALI-2108 at 43 mg/kg and PALI-0008 was administered QD to dogs for 1 day and monitored for key clinical adverse events including emesis (Figure 8).

Figure 1. Diagram depicting how CETSA assesses on-target binding by detecting changes in protein thermal stability upon ligand binding.

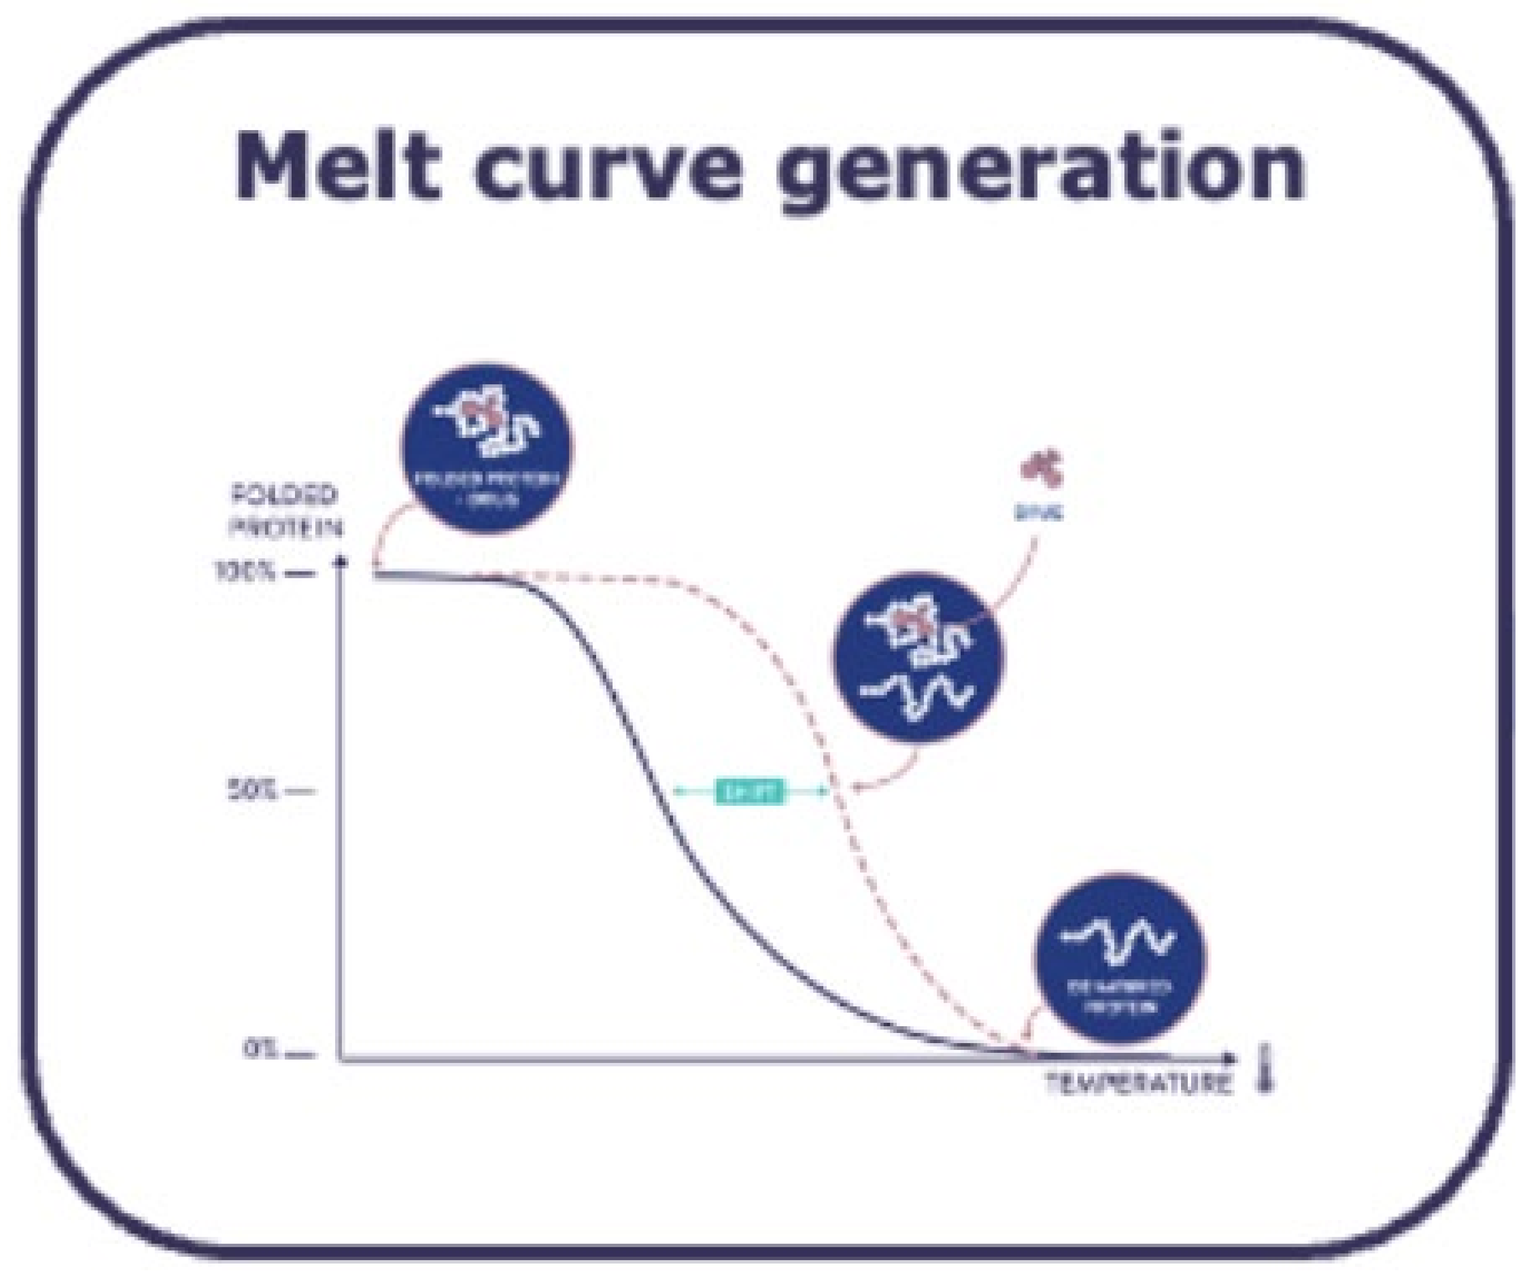

Melt curve generation
FOLDED PROTEIN
100%
50%
0%
TEMPERATURE

Figure 2. Study design of DSS acute colitis disease model utilized to assess PALI-2108 dose dependent efficacy compared to Vehicle control, systemically delivered PDE4 inhibitor apremilast and the immunosuppressant tacrolimus.

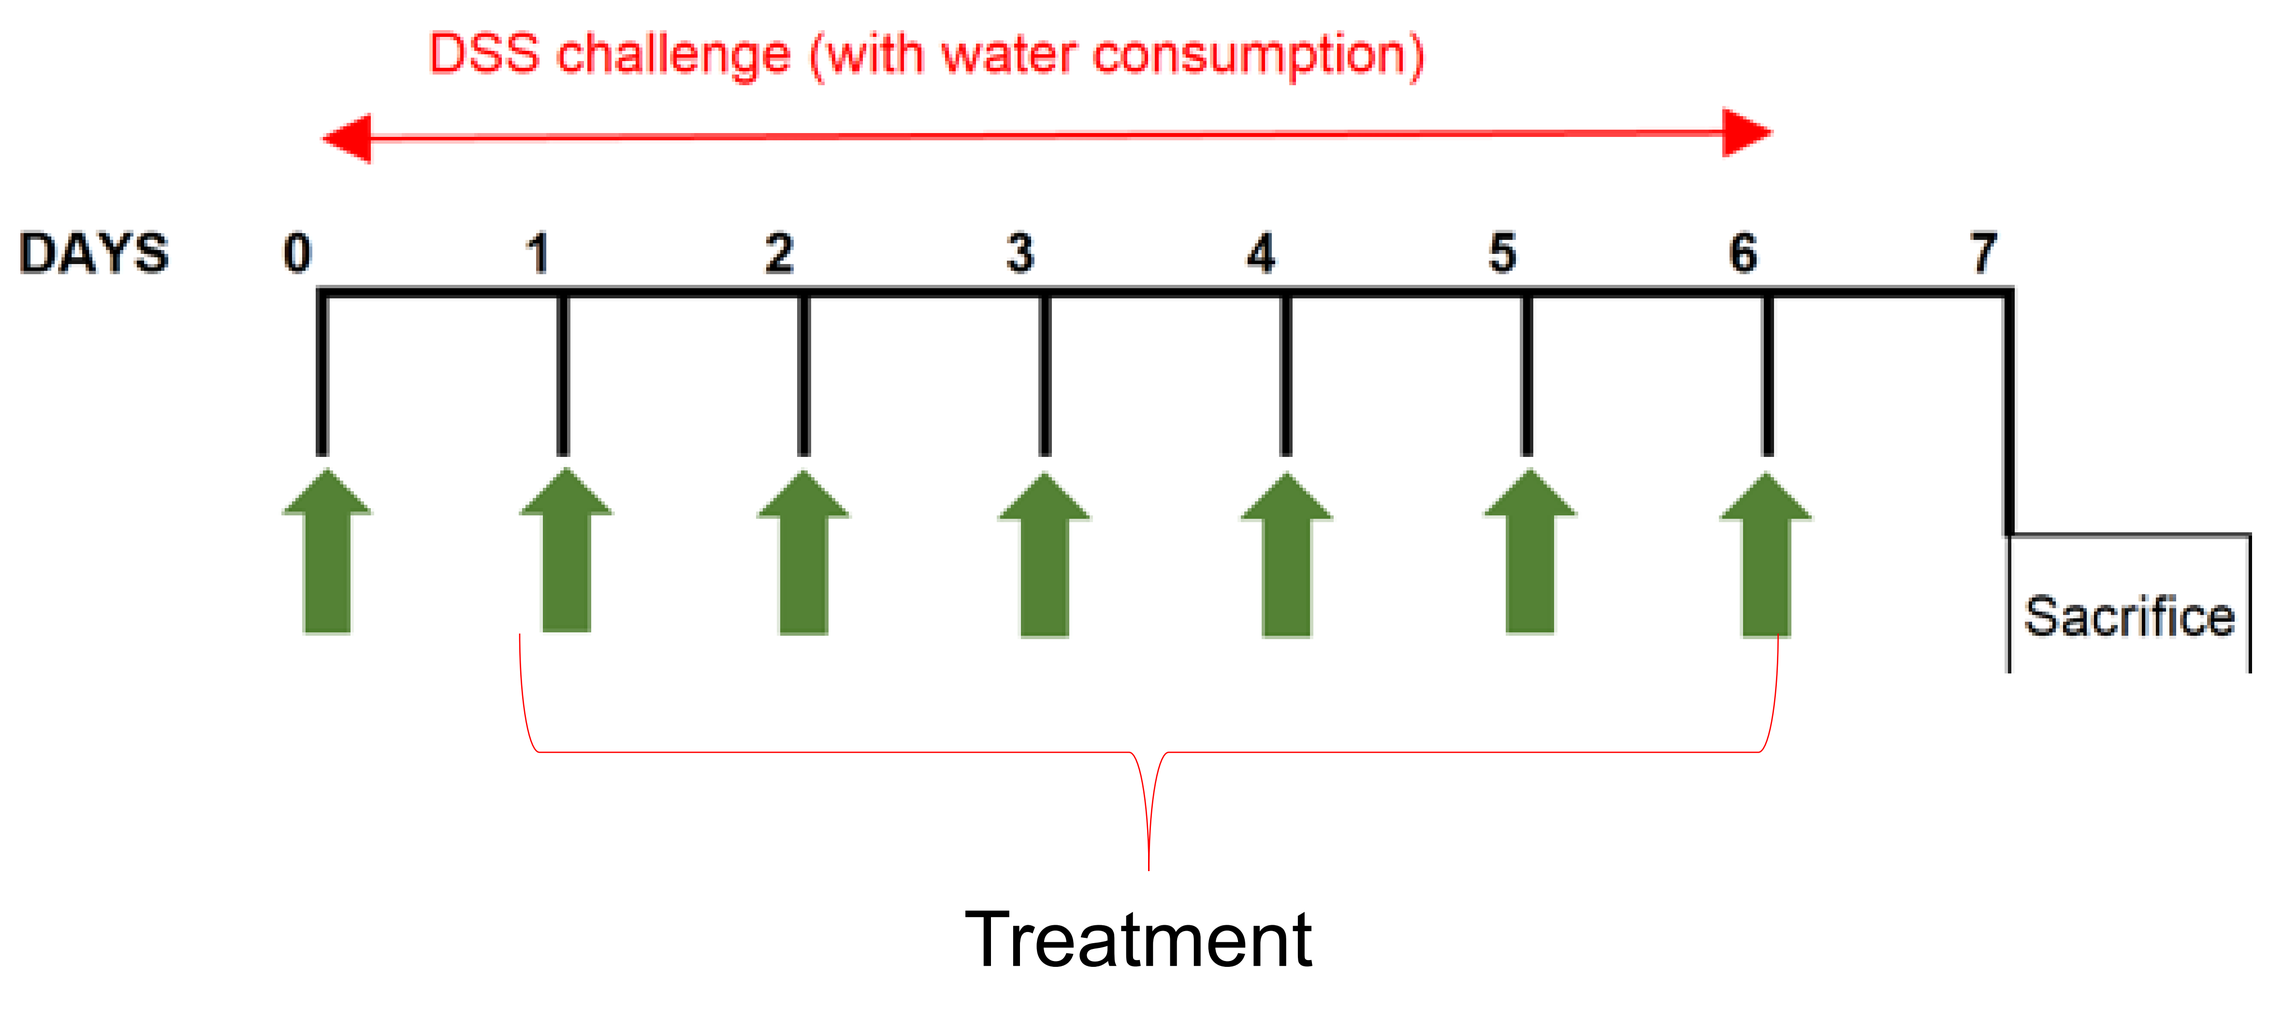

DSS challenge (with water consumption)
DAYS
0
1
2
3
4
5
6
7
Sacrifice
Treatment

Results

Figure 3. PALI-0008 levels in healthy mice dosed PO with PALI-2108. PALI-0008 is favorably distributed to the colon as indicated by a colon/plasma AUC ratio of >200 and the bioavailability of PALI-0008 when delivered as the prodrug PALI-2108 is only 4%

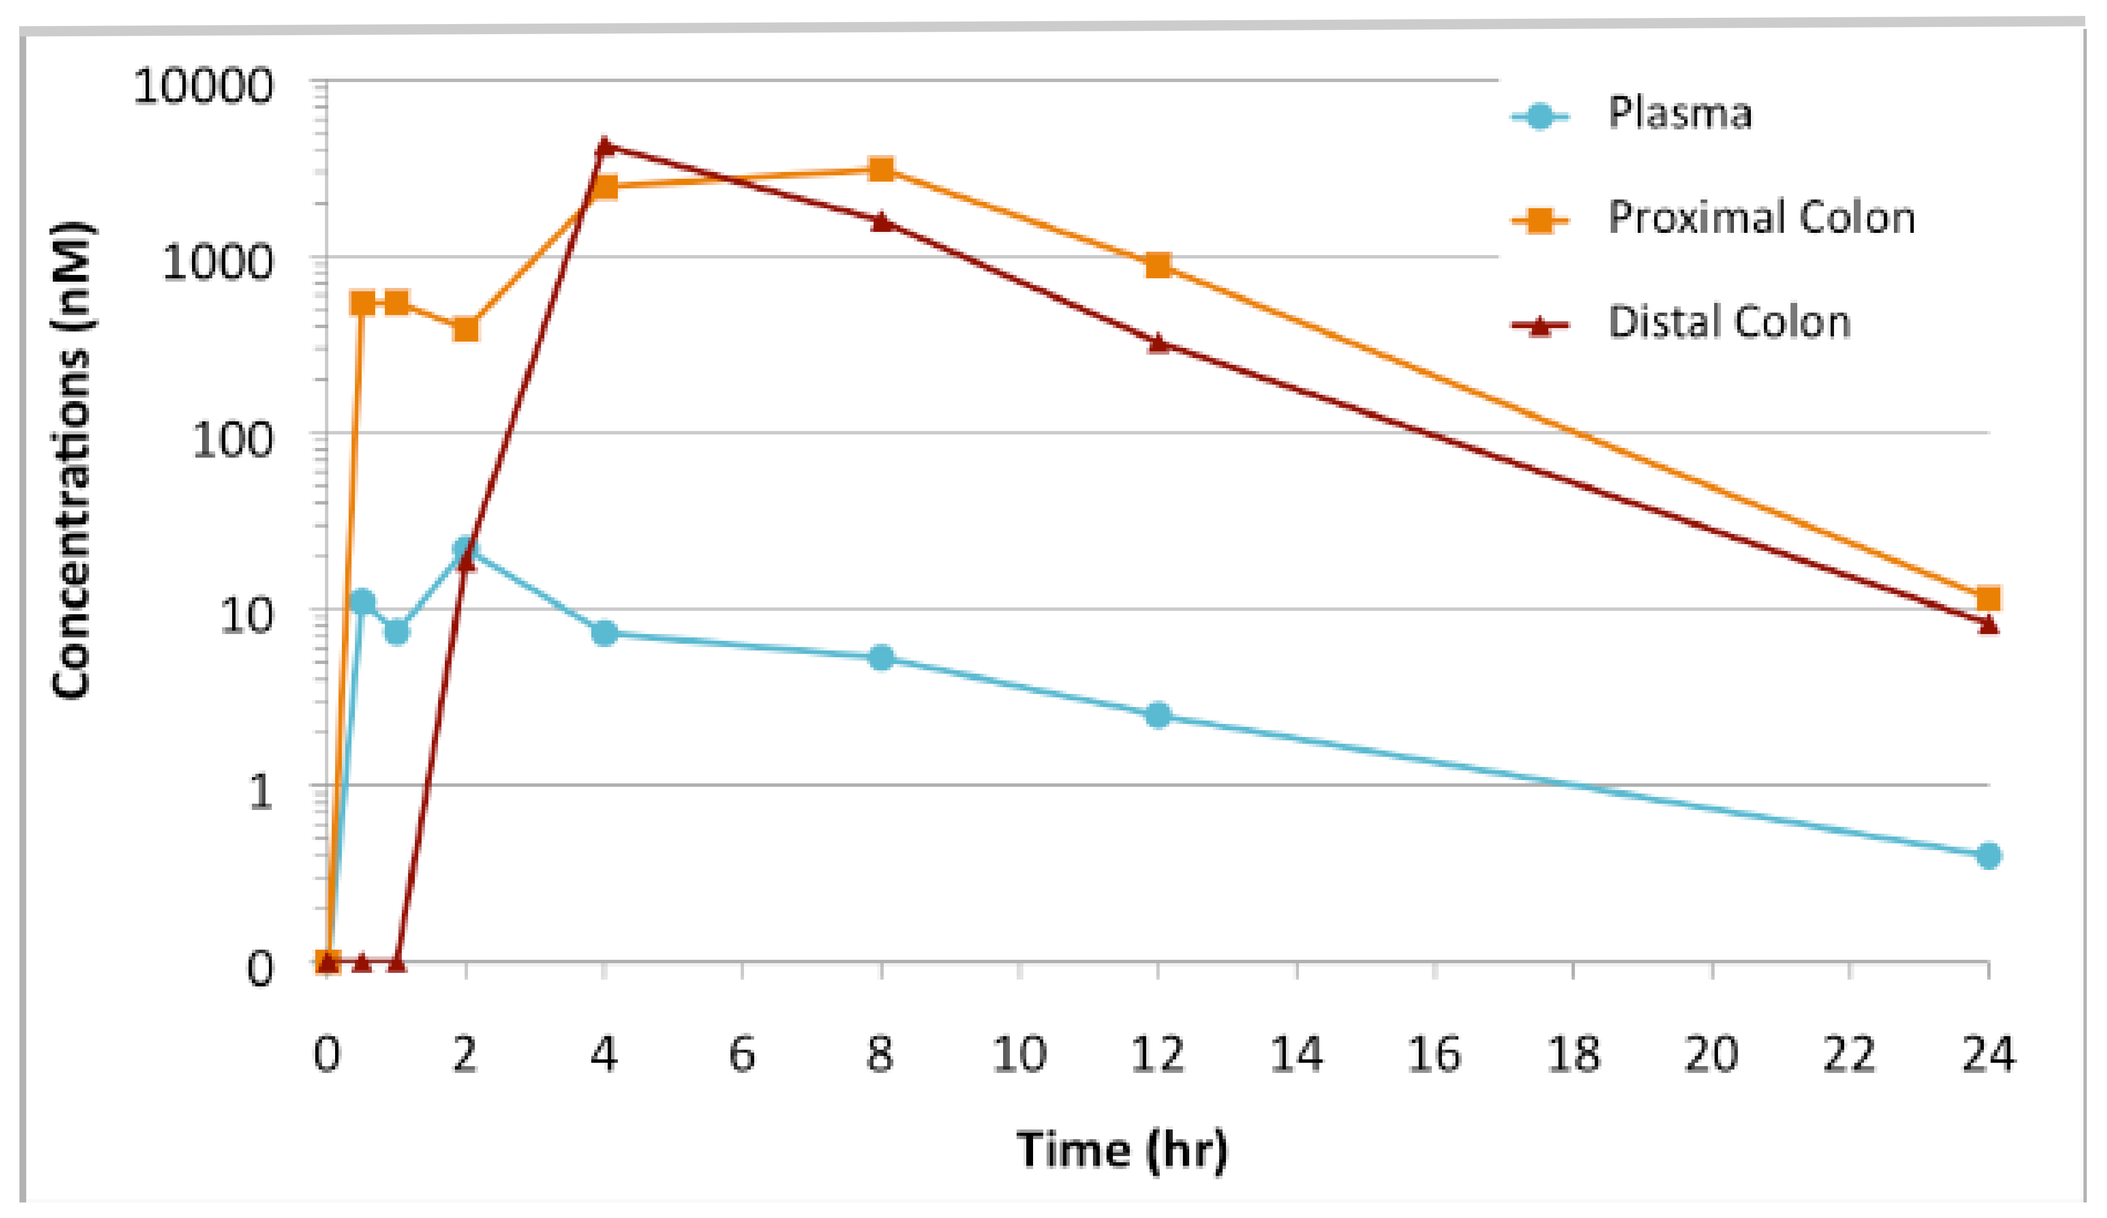

Plasma
Proximal Colon
Distal Colon
Concentrations (nM)
Time (hr)

Figure 4. Concentrations of PALI-2108 (prodrug), PALI-0708 (inactive intermediate) and PALI-0008 (active) where measured at 72h upon dosing PALI-2108 BID at 4.2 mg/kg for 3 days in mice treated once with 3% oxazolone i.r.. Study demonstrated colon bioactivation of PALI-2108 to PALI-0008 and negligible plasma levels of all forms.

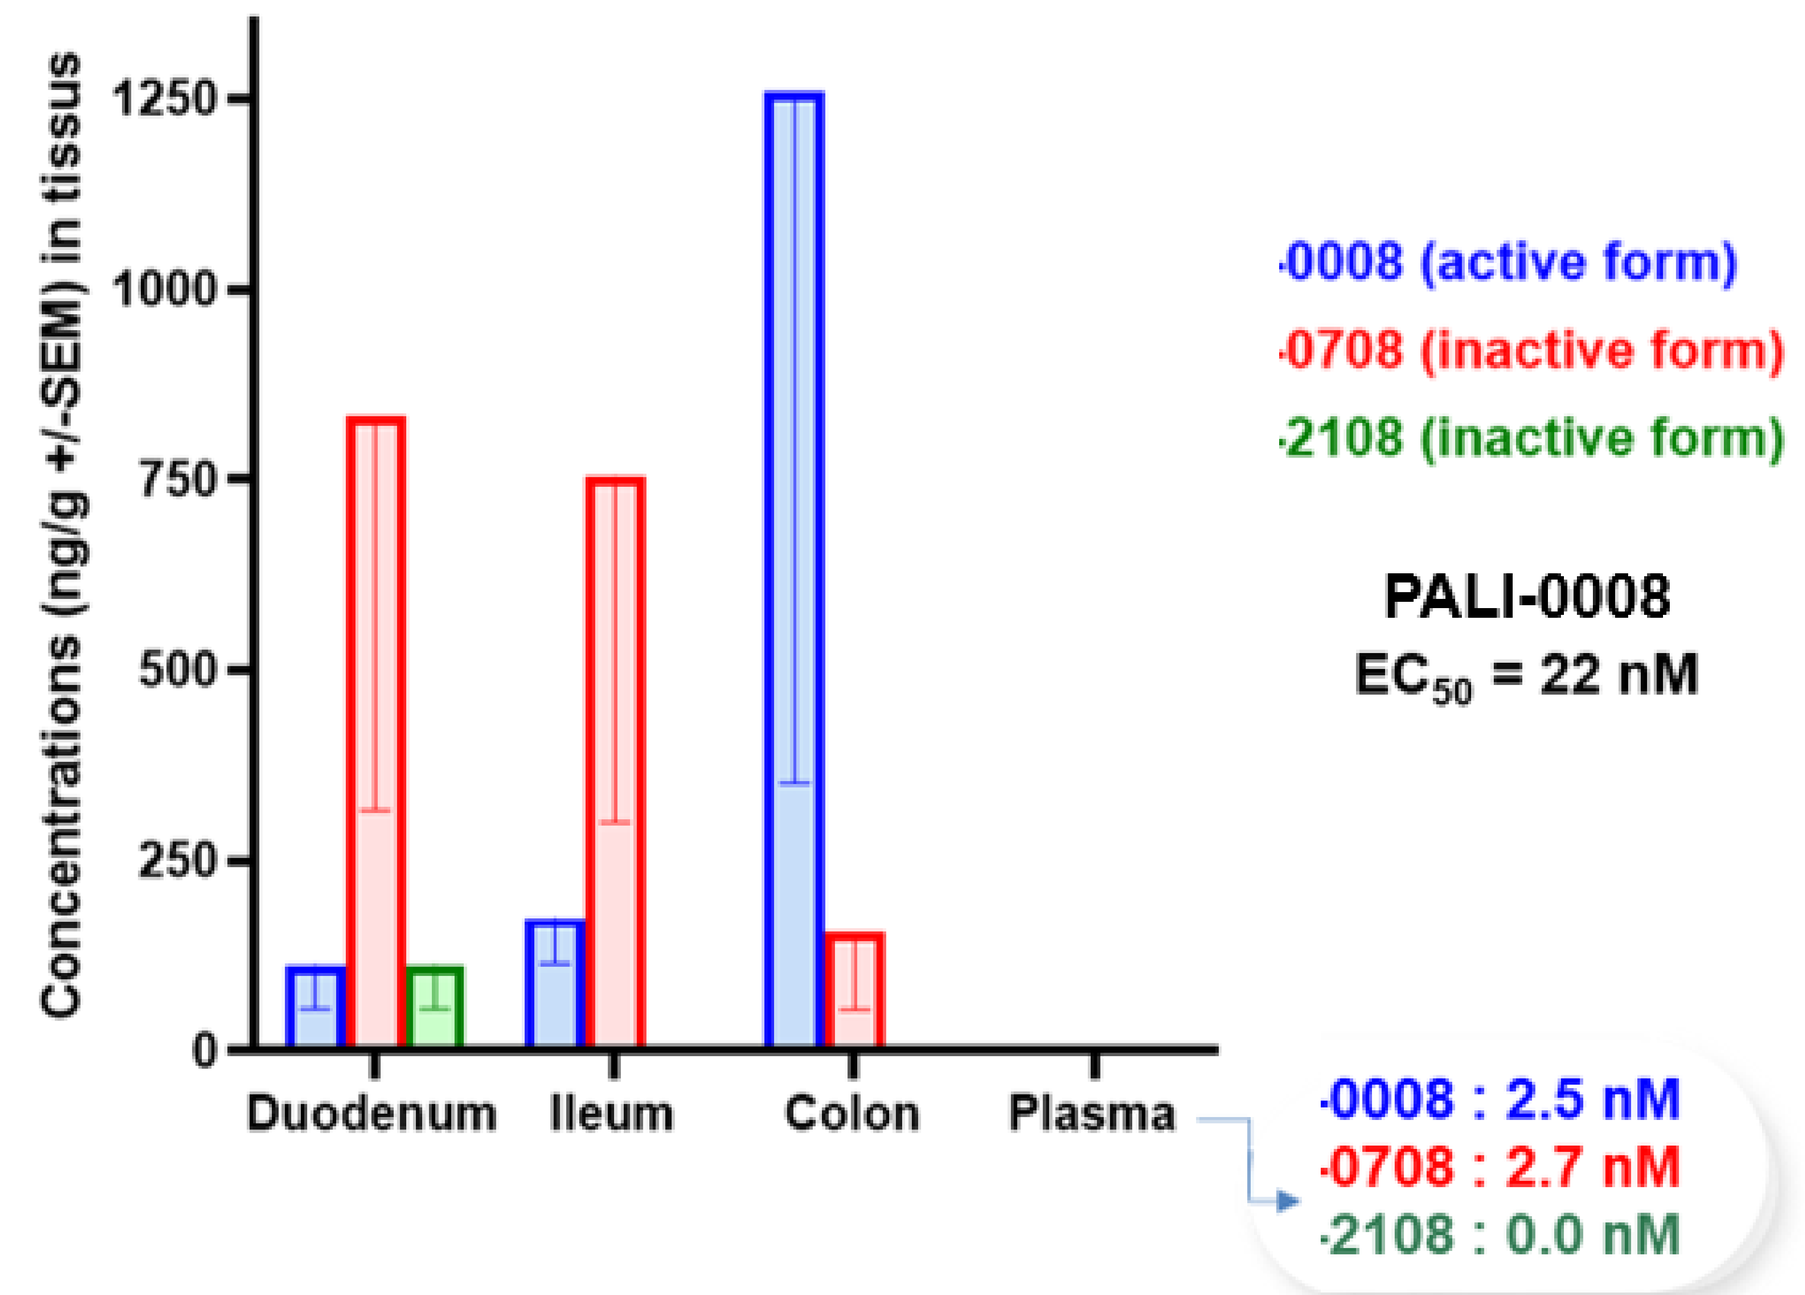

Concentrations (ng/g +/-SEM) in tissus
Duodenum
Ileum
Colon
Plasma
-0008 (active form)
-0708 (inactive form)
-2108 (inactive form)
PALI-0008
EC50 = 22 nM
-0008 : 2.5 nM
-0708 : 2.7 nM
-2108 : 0.0 nM

Figure 5. Mouse colon homogenate with 10 µM of apremilast resulted in a stable assay for PDE4D where the target protein was thermally stabilized by the compound. Target engagement was validated in tissue from animals that were in vivo dosed with apremilast, PALI-2108 or Vehicle controls. For both compounds a thermal stabilization effect on PDE4D was observed confirming target engagement in vivo.

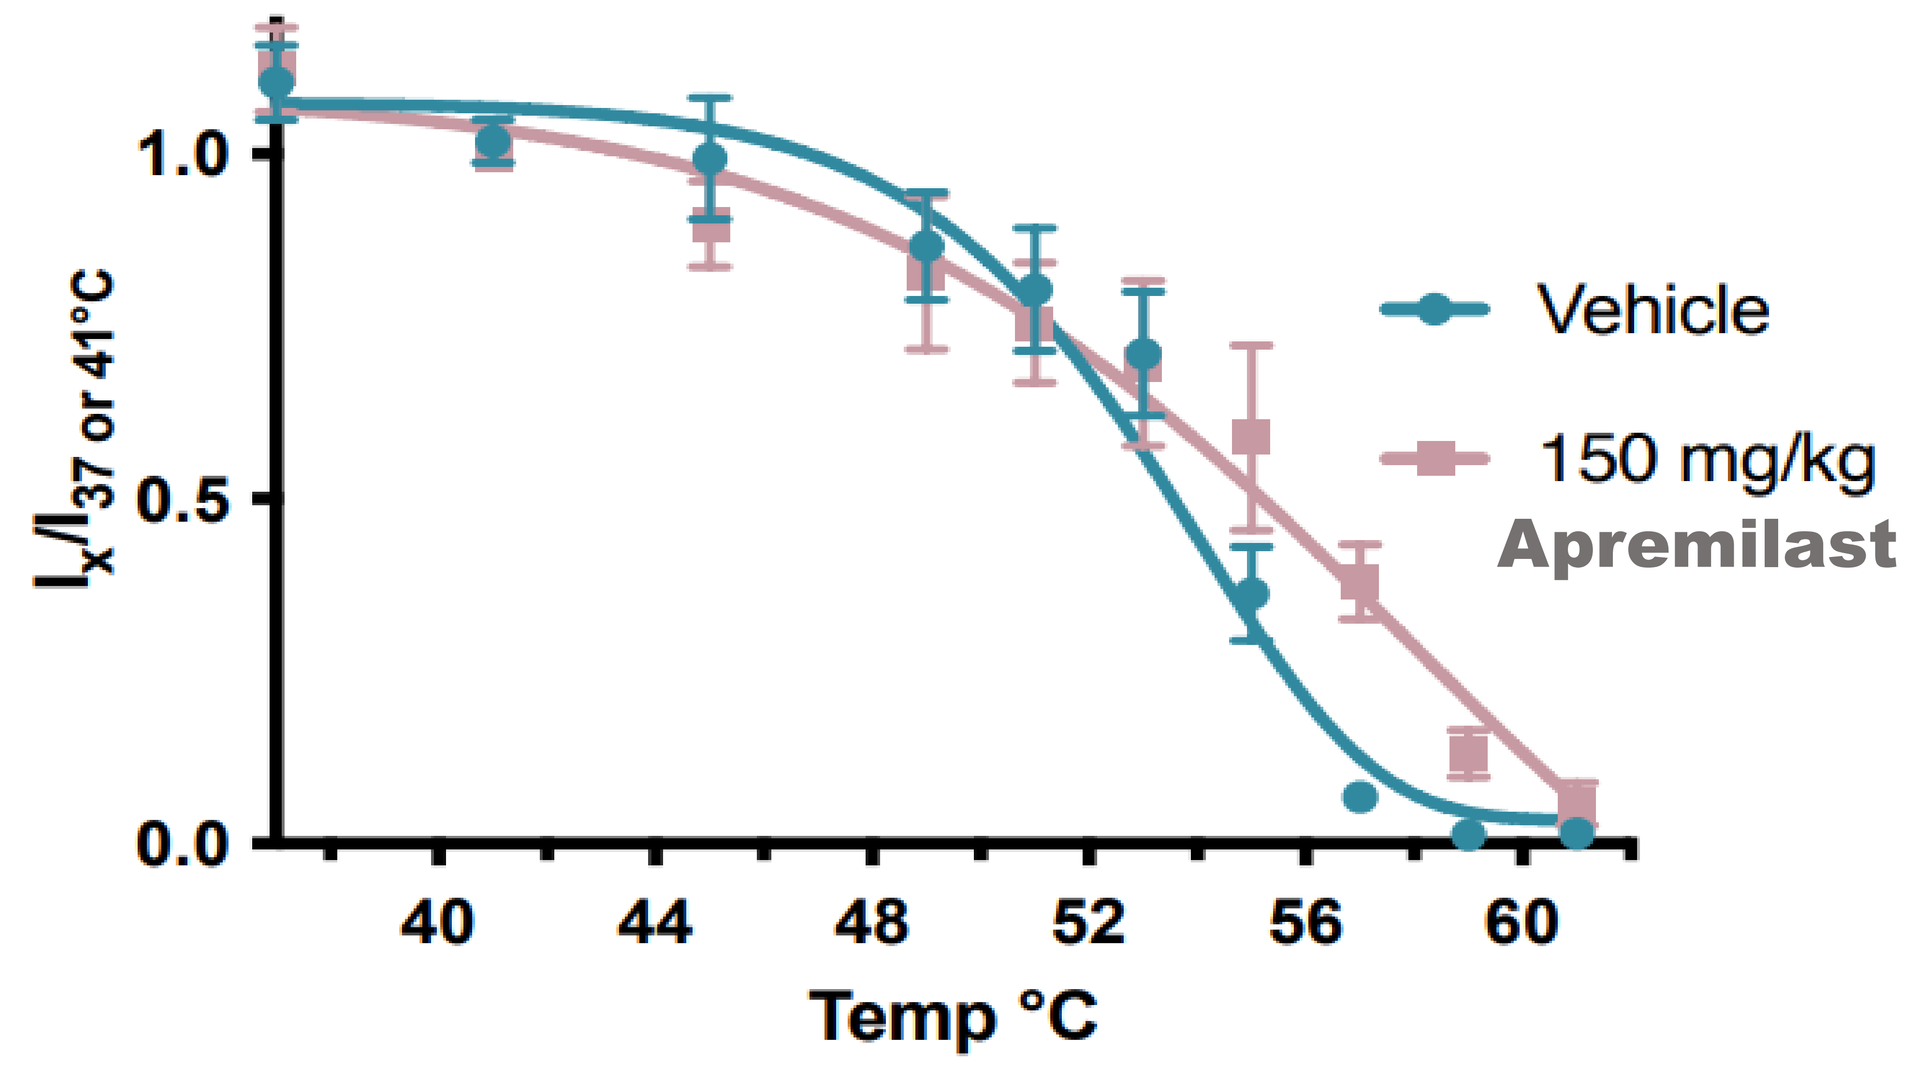

Vehicle
150 mg/kg
Apremilast
Temp °C

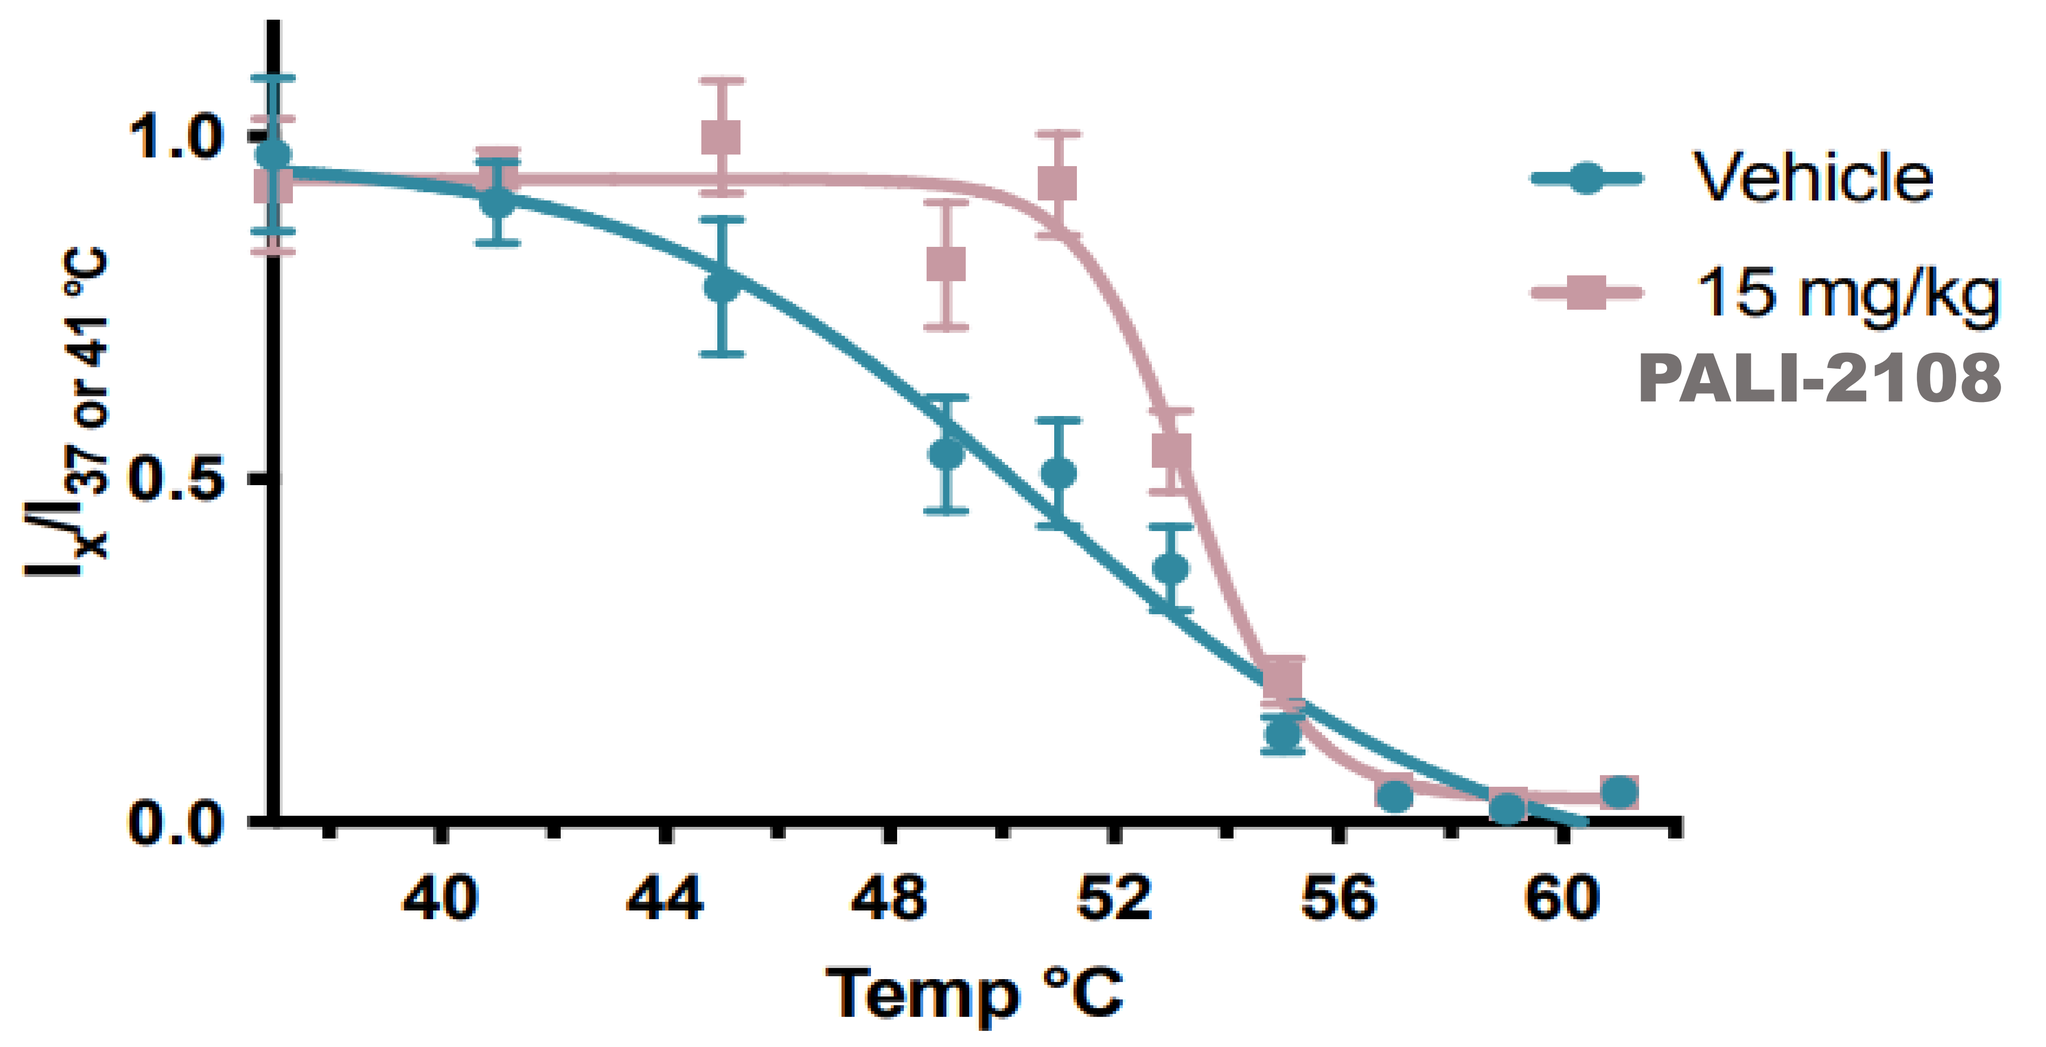

Vehicle
15 mg/kg
PALI-2108
Temp °C

Figure 6. Disease activity index (DAI) score was assessed based on a scoring system including body weight loss, stool consistency and the presence of blood in feces in DSS acute colitis mouse model. Treatment with PALI-2108 at 21, 4 and 0.75 mg/kg BID and 1.5 mg/kg daily significantly decreased the mean DAI score compared to DSS control.

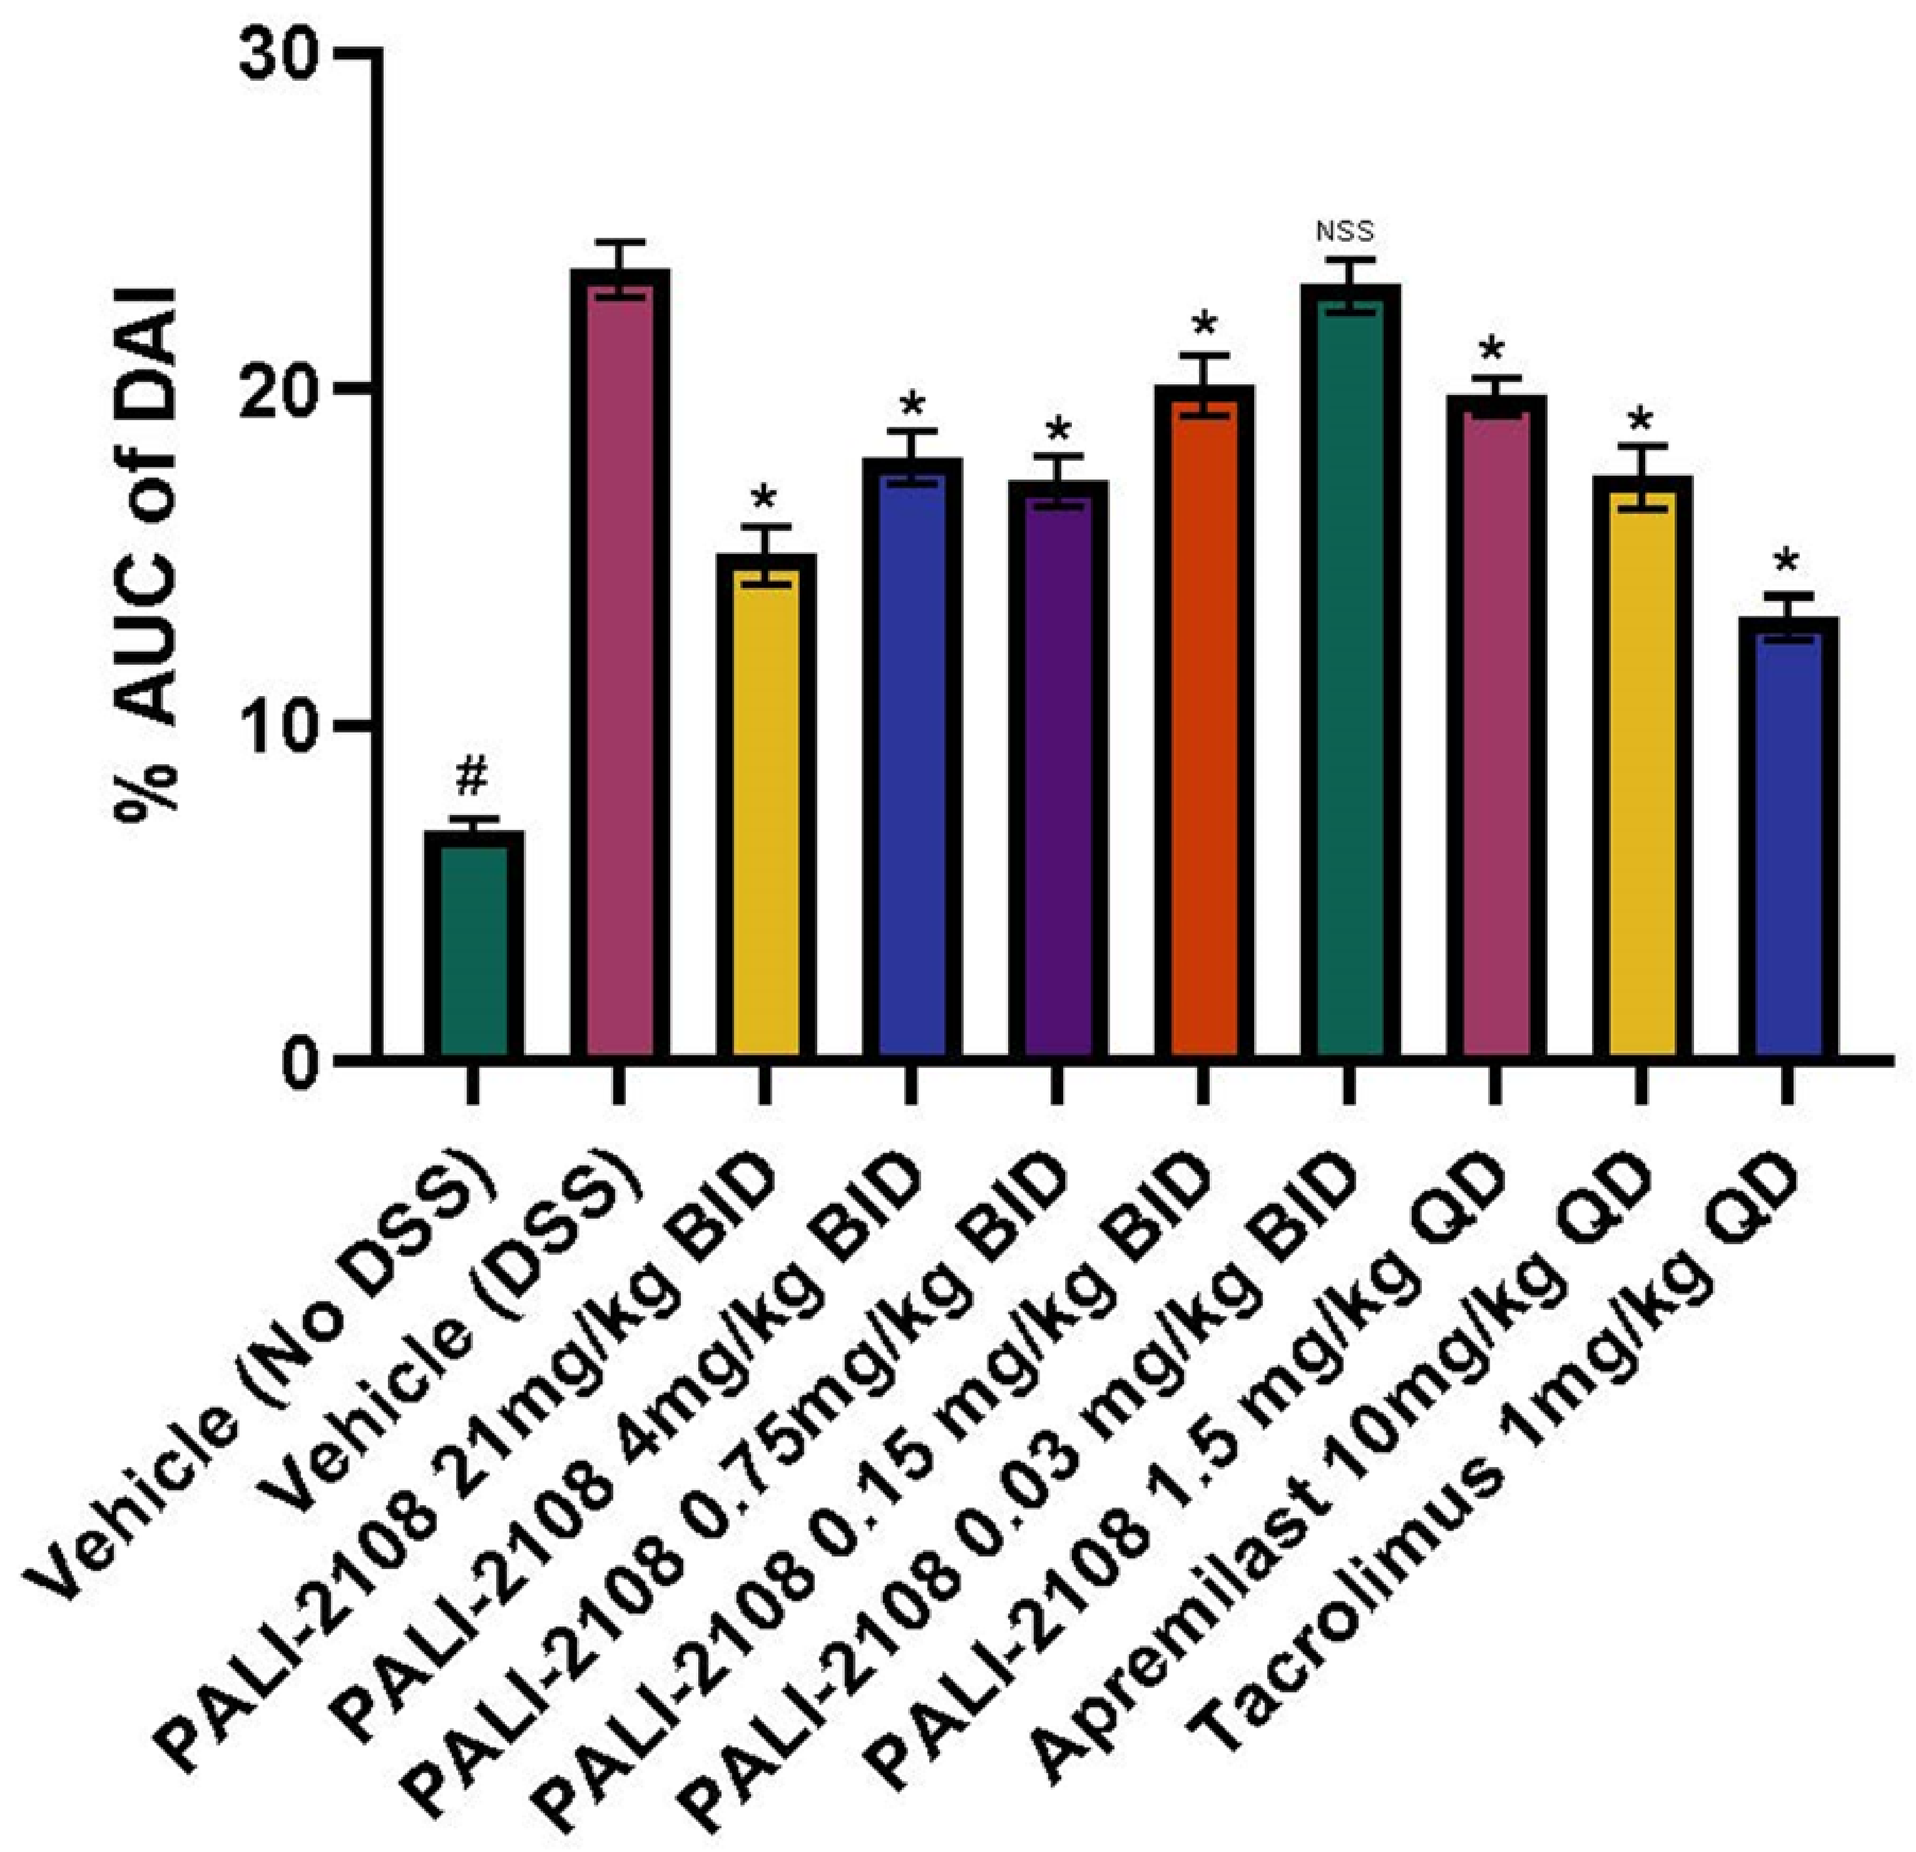

% AUC of DAI
Vehicle (No DSS)
Vehicle (DSS)
PALI-2108 21mg/kg BID
PALI-2108 4mg/kg BID
PALI-2108 0.75mg/kg BID
PALI-2108 0.15 mg/kg BID
PALI-2108 0.03 mg/kg BID
PALI-2108 1.5 mg/kg QD
Apremilast 10mg/kg QD
Tacrolimus 1mg/kg QD
NSS

Figure 7. Treatment with PALI-2108 at 21, 4 and 0.75 mg/kg BID and 1.5 mg/kg decreased the mean body weight loss score for DAI compared to DSS Vehicle control group in DSS mouse model.

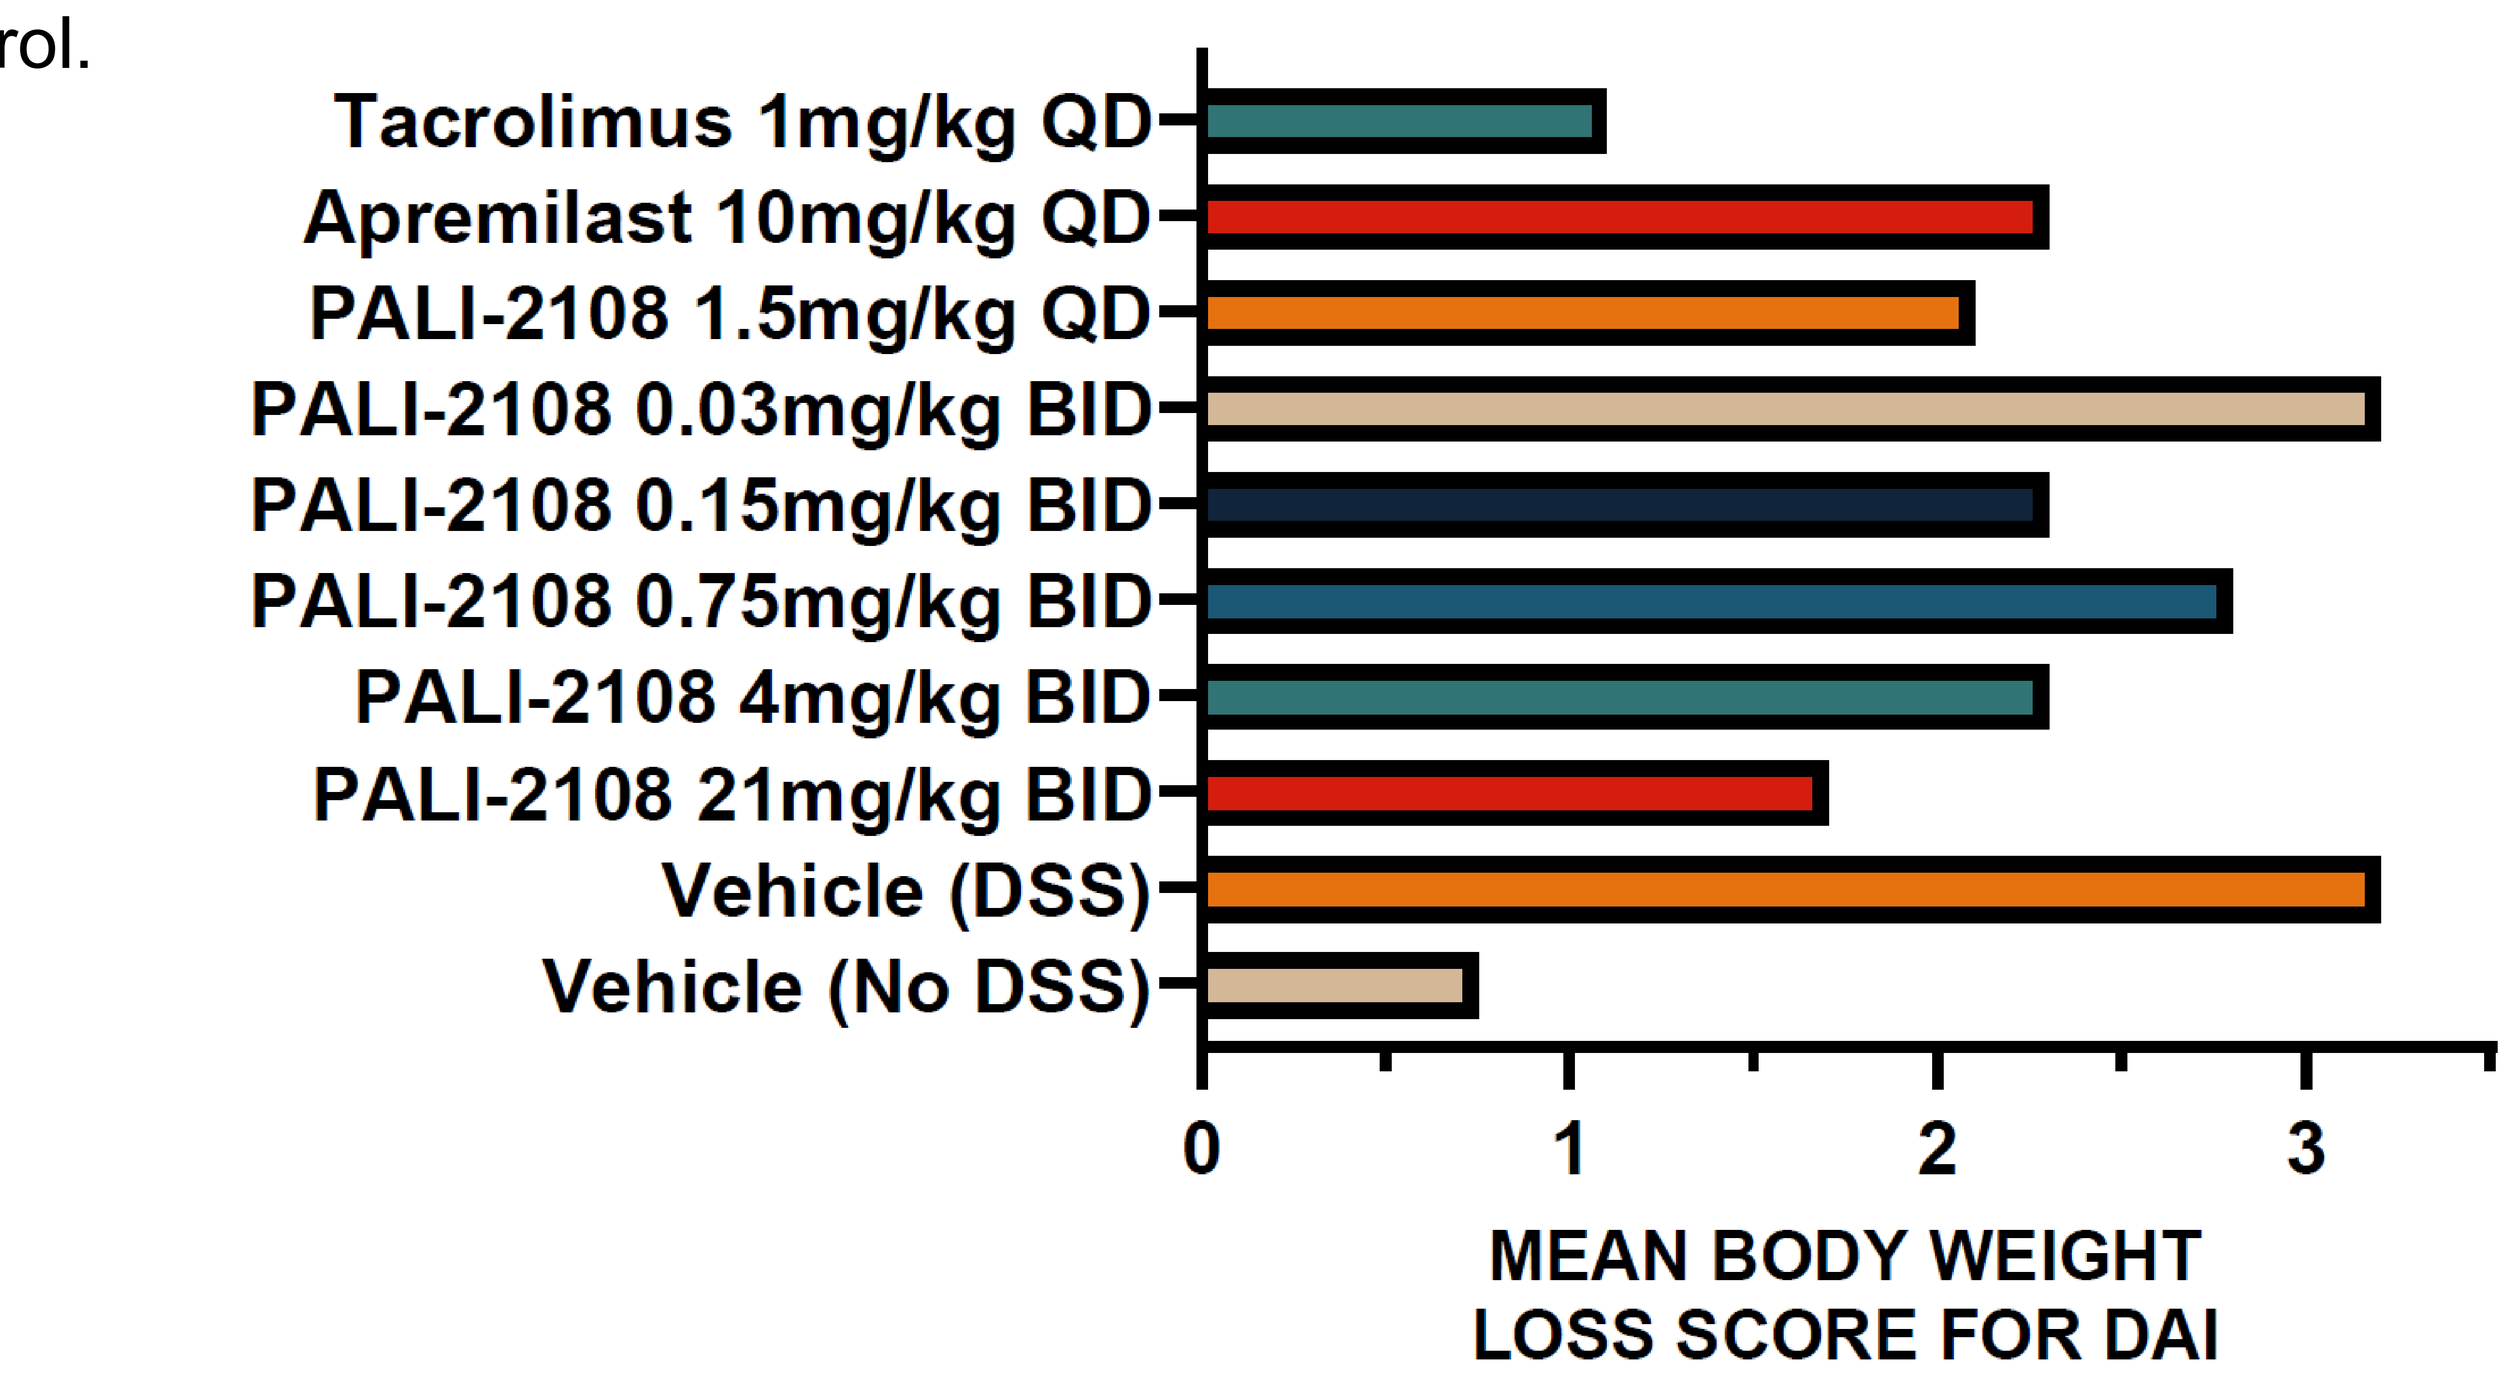

Tacrolimus 1mg/kg QD
Apremilast 10mg/kg QD
PALI-2108 1.5mg/kg QD
PALI-2108 0.03mg/kg BID
PALI-2108 0.15mg/kg BID
PALI-2108 0.75mg/kg BID
PALI-2108 4mg/kg BID
PALI-2108 21mg/kg BID
Vehicle (DSS)
Vehicle (No DSS)
MEAN BODY WEIGHT LOSS SCORE FOR DAI

Figure 8. PO administration of prodrug PALI-2108 at 43 mg/kg was well tolerated, while PO administration of PALI-0008 resulted in emesis at 1 and 3 mg/kg, suggesting systemic toxicity only with administration of the active compound and not the prodrug.

Species	Route of admin	Compound	Dose (mpk)	Analysed compound	C_{max} (nM)	Emesis
Dog	PO; QD	PALI-2108	43	PALI-2108	11	NO
				PALI-0008	28	
				PALI-0708	9	
Dog	PO; QD	PALI-0008	0.1	PALI-0008		NO
			0.3			NO
			1		139	Yes 2/3
			3		270	Yes 3/3

Figure 9. PALI-2108 mechanism of action. PDE4 is highly expressed in immune and proinflammatory cells. The inhibition of PDE4 prevents transcription factors, such as NF-κB, from inducing the expression of various proinflammatory mediators involved in IBD, such as TNF-α and various interleukins. The PDE4 inhibitor PALI-2108 is orally delivered and colon activated allowing for local activity with low systemic exposure.

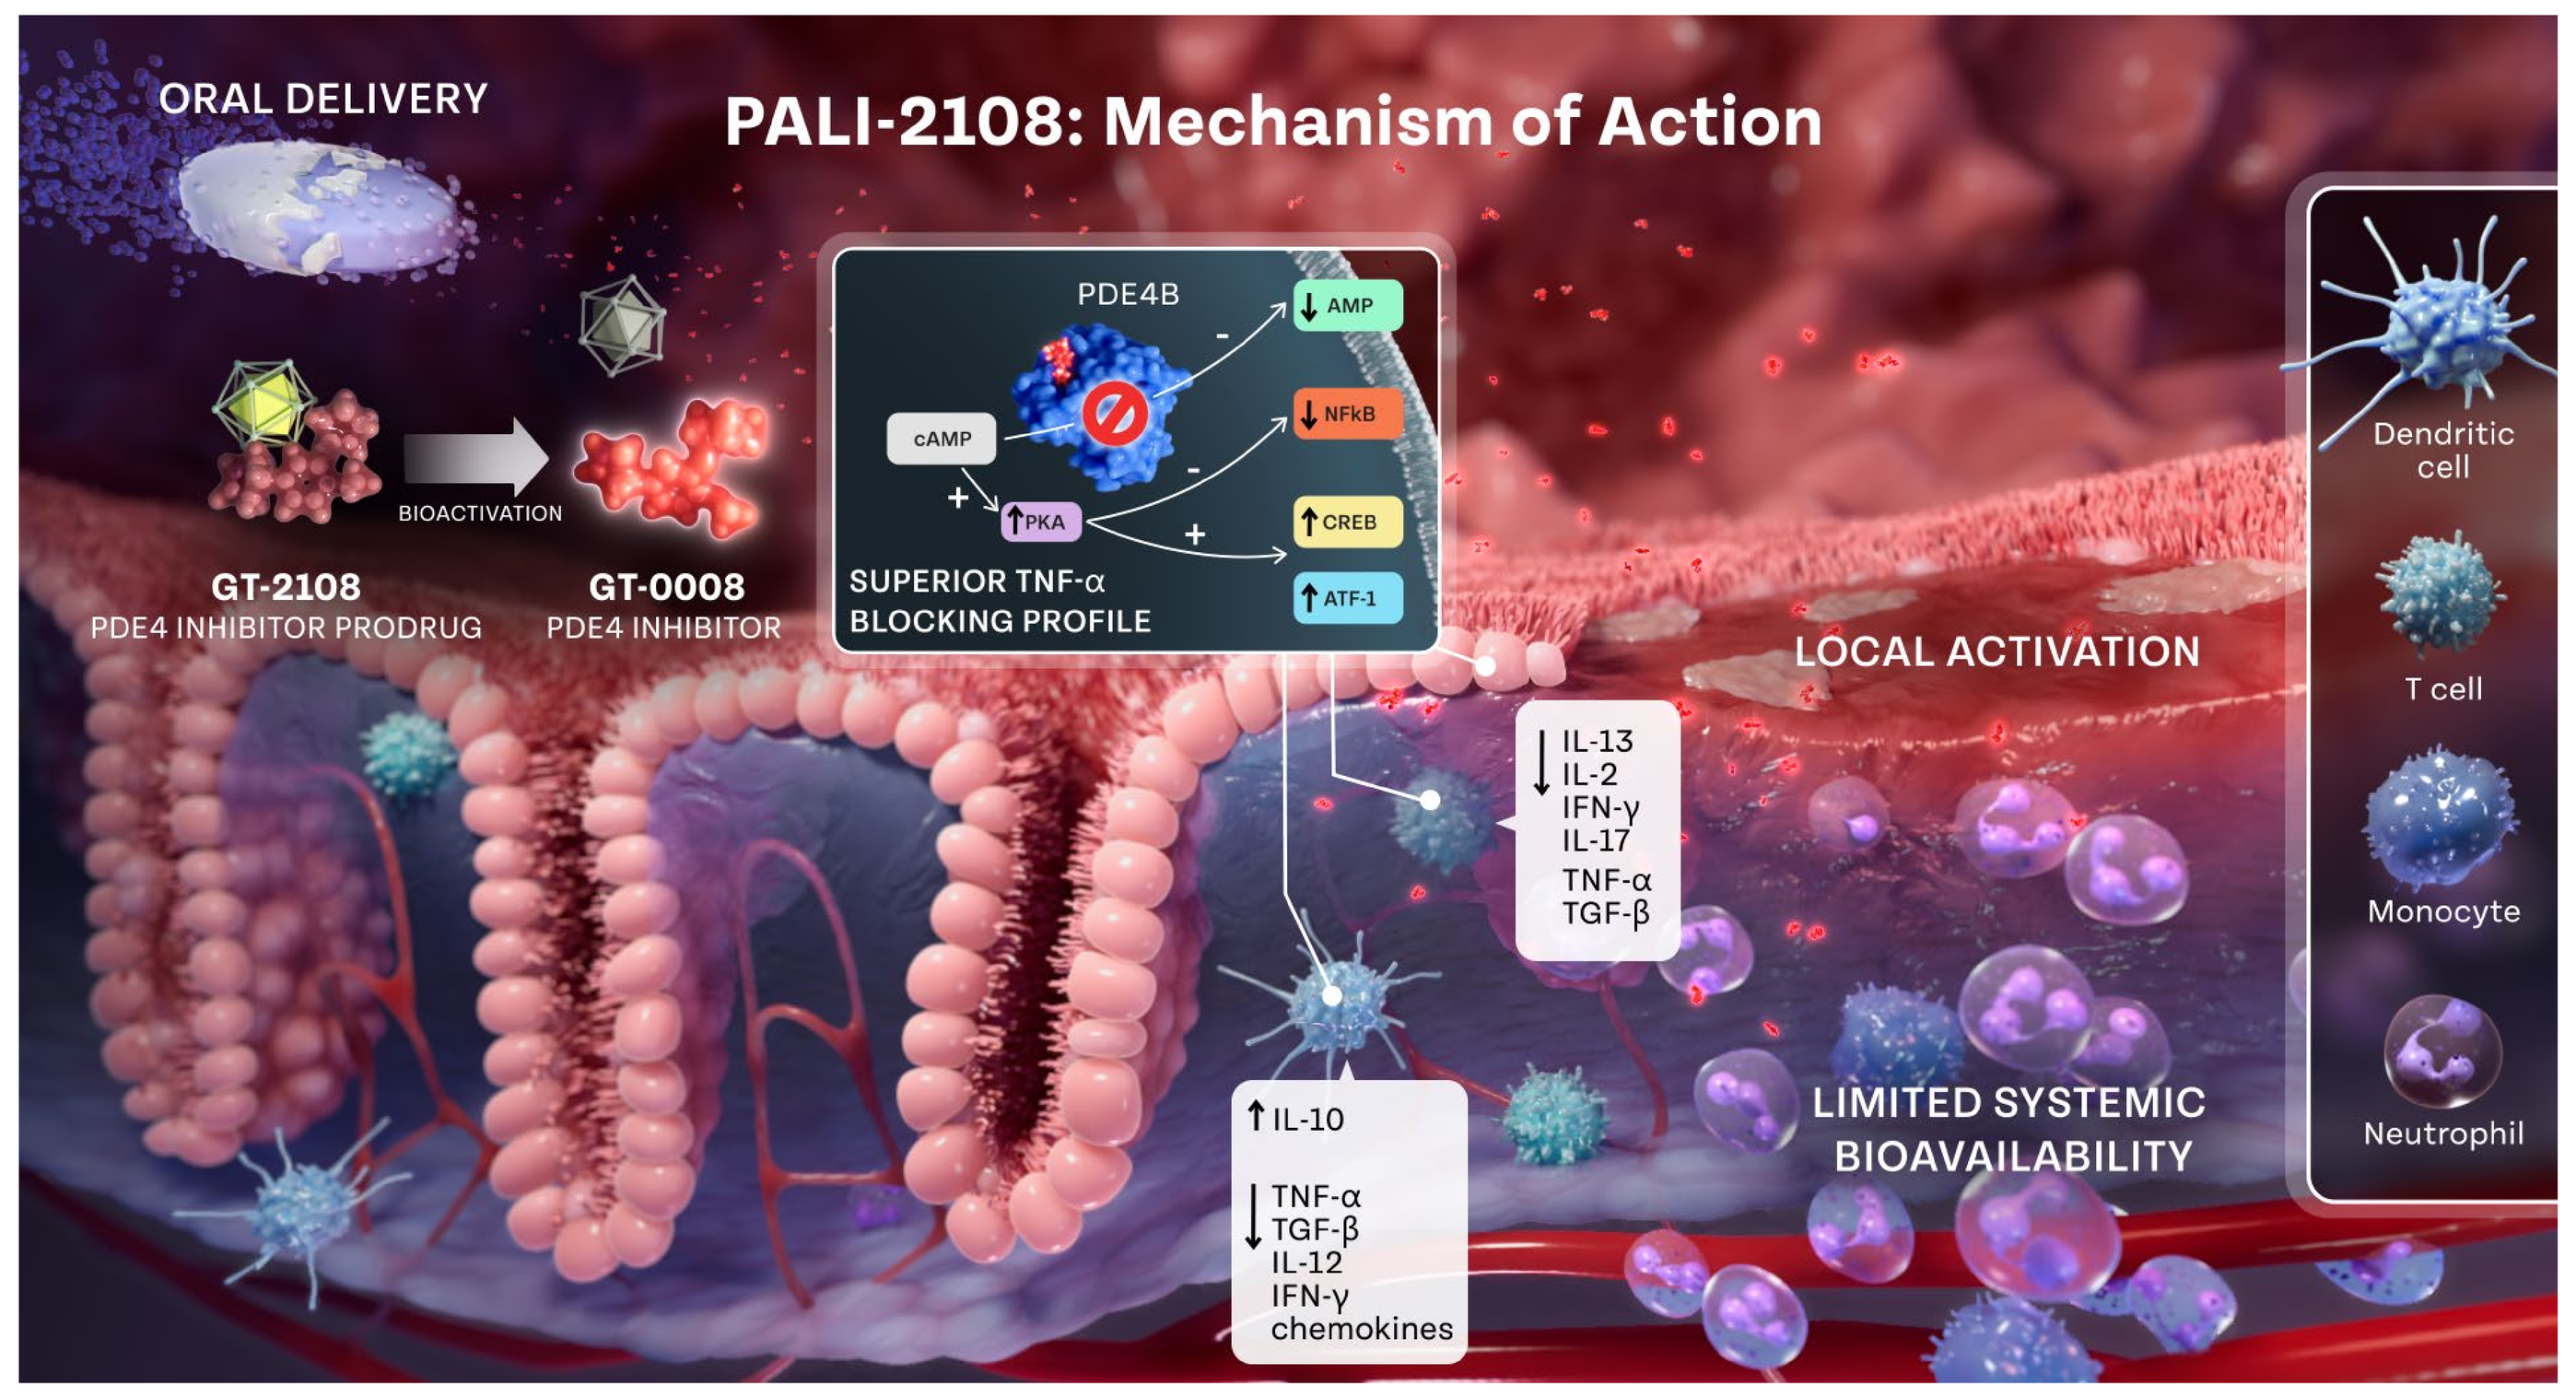

ORAL DELIVERY
PALI-2108: Mechanism of Action
GT-2108
PDE4 INHIBITOR PRODRUG
BIOACTIVATION
GT-0008
PDE4 INHIBITOR
PDE4B
SUPERIOR TNF-α BLOCKING PROFILE
LOCAL ACTIVATION
LIMITED SYSTEMIC BIOAVAILABILITY
Dendritic cell
T cell
Monocyte
Neutrophil

Conclusions

PDE4 inhibitor pro-drug, PALI-2108 shows:

- Local bioactivation
- Colon-specific distribution
- Similar target engagement to other PDE4 inhibitors
- Dose-dependent efficacy in a mouse UC model
- No systemic toxicity in dog and large therapeutic window due to local activation

PALI-2108 is a promising novel therapy for UC, with localized bioactivation, expanded therapeutic window, and potent PDE4 inhibitory activity. PALI-2108 is in development for moderate-to-severe UC and is advancing toward regulatory filing for first-in-human studies.

CONTACT INFO
Joie Bernard
Palisade Bio
Carlsbad, CA
joie.bernard@palisadebio.com